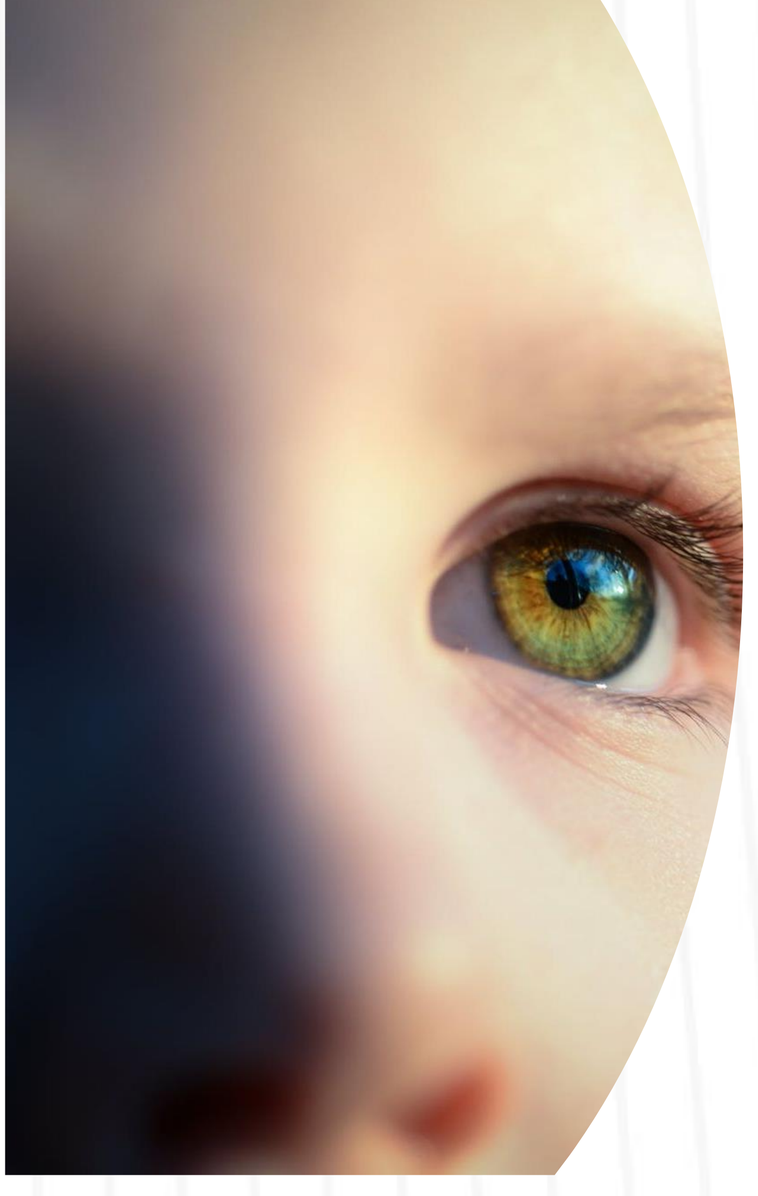

The PLATYPUS Study: A Phase 1 First-in-Human Study of VP-001, a peptide-oligonucleotide conjugate designed to treat Retinitis Pigmentosa Type 11 (RP11) due to pathogenic *PRPF31* variants

Fred K. Chen[1,2], David G. Birch[4], Carla Jackson[3], Anna Mills[3], Ianthe Pitout, Laura Florez[3], Jessica Nichols[3], Paula Cunningham[3], Janya Grainok[3], Adam Martin[3], Subrata Das[3], Sue Fletcher[3], George Mitchell[3], Clare Guerrero[3], Sri Mudumba[3]

[1]Centre for Ophthalmology and Visual Science, The University of Western Australia, Perth, Western Australia, Australia; [2]Ocular Tissue Engineering, Lions Eye Institute, Nedlands, Western Australia, Australia; [3]PYC Therapeutics, Perth, Western Australia, Australia; [4]Retina Foundation of the Southwest, Dallas, Texas, United States

RP11 is a progressive, blinding eye disease of childhood for which there are no available treatments

Retinitis Pigmentosa type 11 (RP11) & VP-001

- Dominantly inherited, severe and progressive **blinding eye disease** that begins in childhood
- Retinitis Pigmentosa type 11 affects ~1 in 100,000 people[1-11]
- There are no treatments currently available nor are there any in clinical development
- **RP11 is a monogenic disease**: 2-5x higher likelihood of therapeutic success in human studies[12]
- Caused by **haploinsufficiency of *PRPF31*** expression in retinal pigment epithelium and photoreceptors
- **VP-001 restores PRPF31 expression** towards levels seen in unaffected individuals

RP11 patient vision deterioration

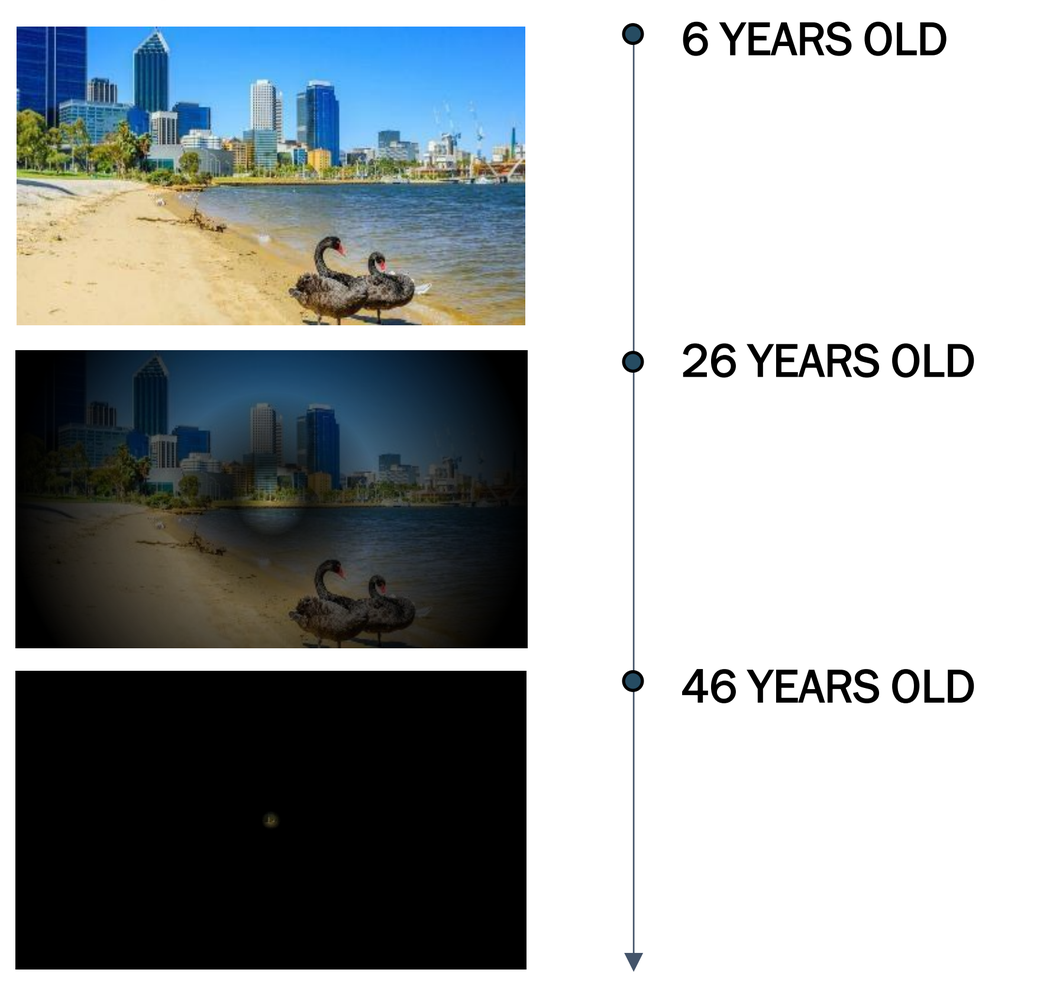

6 YEARS OLD
26 YEARS OLD
46 YEARS OLD

PYC's technology overcomes the delivery challenge of antisense therapies

The primary challenge for genetic medicines is delivering enough drug to the target

RNA modalities are an approved class of drug but their efficacy is limited by an inability to cross the lipid bilayer and reach their intracellular target

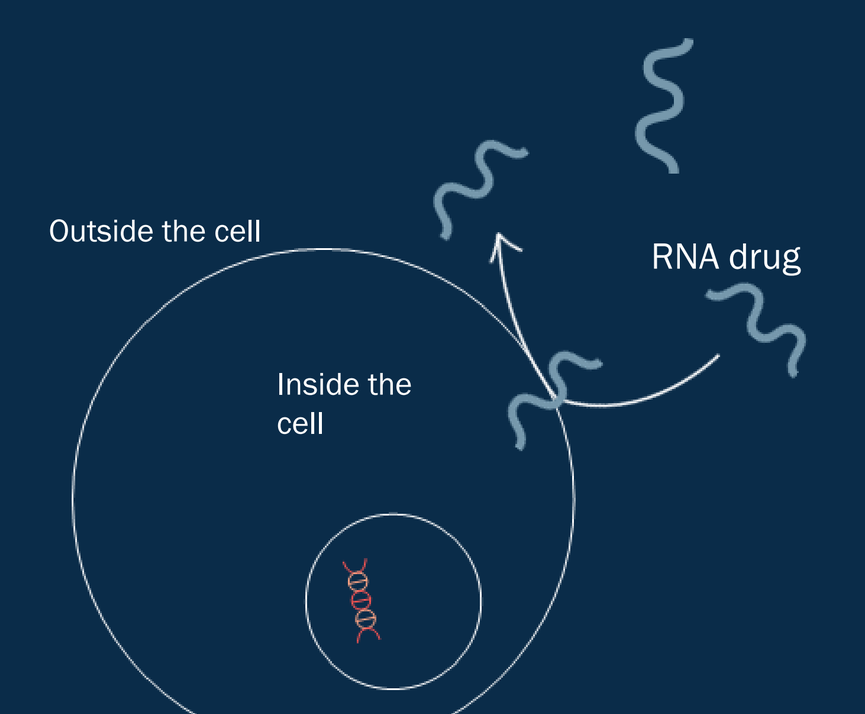

Outside the cell
RNA drug
Inside the cell

PYC's proprietary drug delivery technology facilitates cellular entry for the RNA drug to reach its target

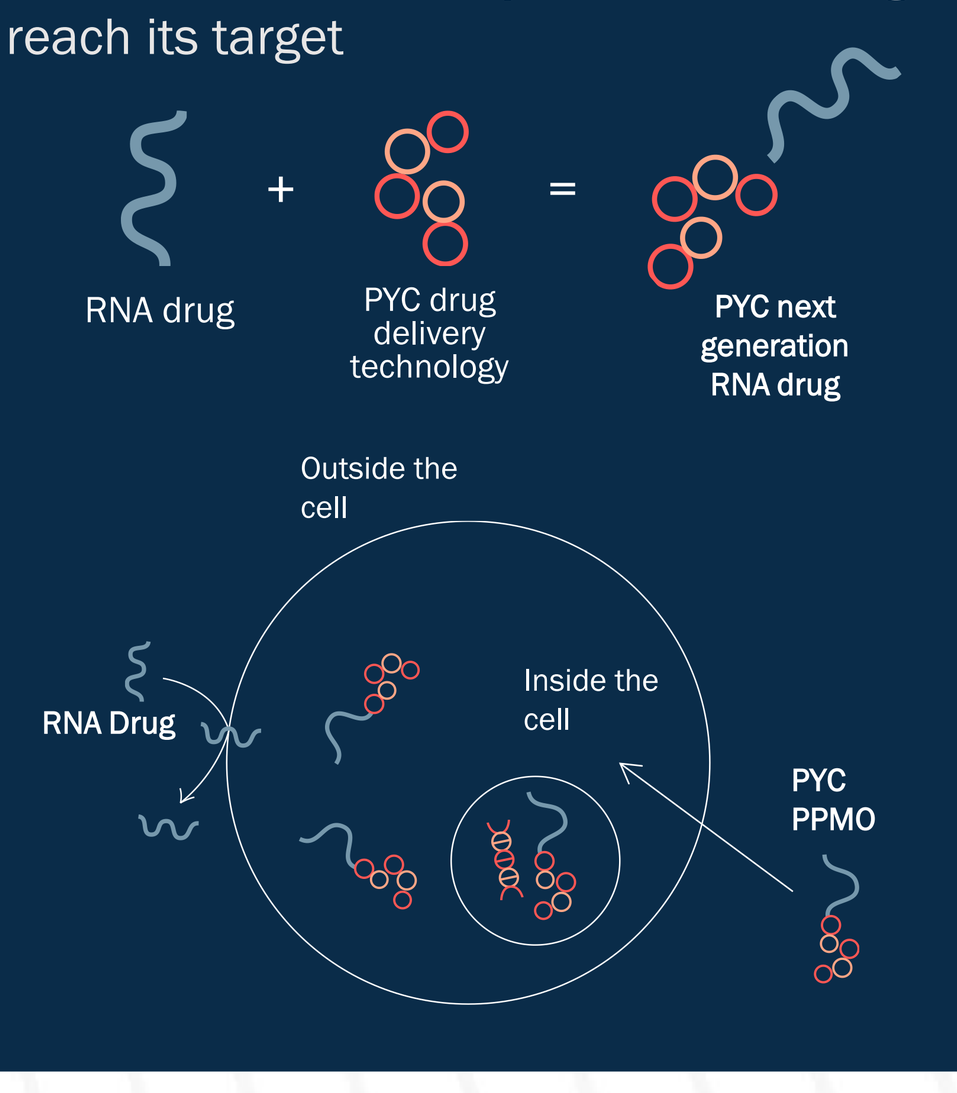

RNA drug
+
PYC drug delivery technology
=
PYC next generation RNA drug
Outside the cell
Inside the cell
RNA Drug
PYC PPMO

Antisense Therapies for Retinal Disease

- Antisense oligomers (ASO) are synthetic nucleic acid analogues that can be designed to modify pre-mRNA splicing or protein expression for the treatment of disease
- Suboptimal ASO delivery presents an ongoing challenge and limits the realisation of RNA therapeutics[3]
- Antisense therapies for retinal disease are particularly limited in their ability to achieve adequate cellular uptake in the retina[4]
- PYC's cell-penetrating peptide (CPP) platform facilitates phosphorodiamidate morpholino oligomer (PMO) delivery to the retinal cells affected by RP11

VP-001 is designed to skip exon 17 in *CNOT3* to achieve upregulation of PRPF31 expression

CNOT3 is a negative regulator of *PRPF31*.[13] Modulation of CNOT3 function by ASO-mediated exon skipping enhances *PRPF31* RNA and PRPF31 protein expression

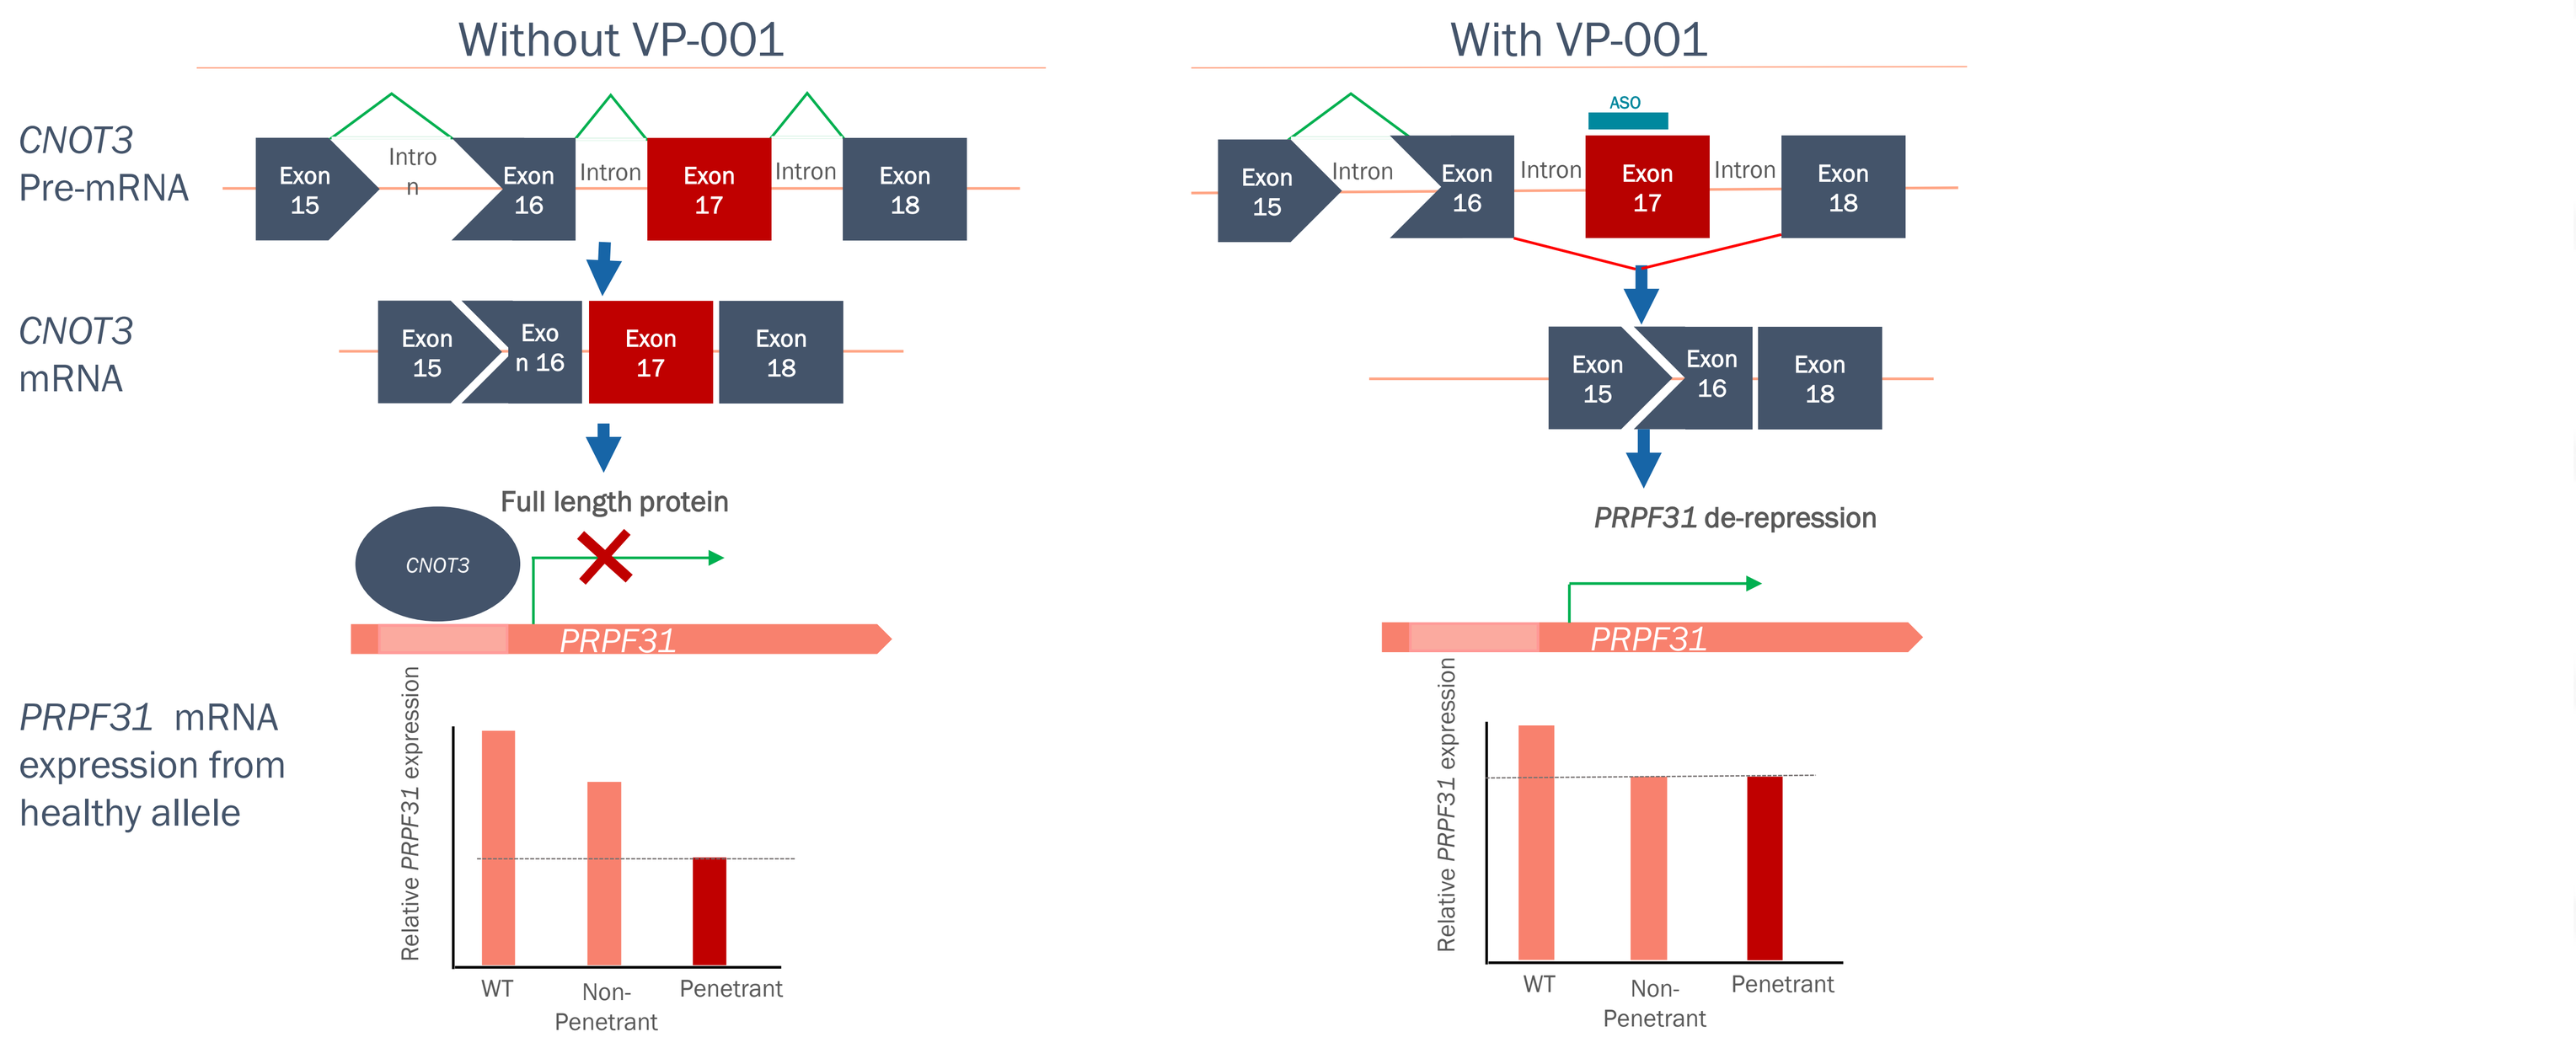

Without VP-001
With VP-001
CNOT3 Pre-mRNA
CNOT3 mRNA
PRPF31 mRNA expression from healthy allele
Exon 15
Intron
Exon 16
Exon 17
Exon 18
ASO
Full length protein
CNOT3
PRPF31
PRPF31 de-repression
Relative PRPF31 expression
WT
Non-Penetrant
Penetrant

Methods

- An exon skipping oligonucleotide conjugated to a cell-penetrating peptide (VP-001) was developed, designed to alter splicing of CNOT3, a negative regulator of *PRPF31*.[13]
- ***In-vitro* preclinical efficacy data** generated in iPSC-derived retinal pigment epithelium (RPE) and 3-dimensional retinal organoids (RO) derived from multiple RP11 patients, confirmed mutation-agnostic intended mechanism of action in target cells, following treatment with VP-001.[14]
- **Ocular Pharmacokinetic and Tolerability study in non-human primates (NHP):** Cynomolgus monkeys were dosed bilaterally with VP-001 by IVT injections of 30 µg/eye . Ocular tissues were dissected from the eyes of two animals after euthanasia at 1, 7, 28, 56, 84 and 112 days post injection.[14]
- **Platypus, a Phase 1 Single Ascending Dose (SAD) First-in-Human (FIH) study:** Twelve participants with *PRPF31*-associated rod-cone dystrophy (RP11) were recruited at 6 sites in the USA in 4 cohorts. One eye of each participant received an intravitreal injection of VP-001 (3, 10, 30 and 75 µg) and followed for 48 weeks for adverse events. Dose escalation was determined by a safety review committee. **Eligible patients were enrolled in an extension study to receive repeat doses of 30 and 75 µg.** Visual acuity, slit lamp examination, microperimetry, fundus autofluorescence imaging, spectral-domain optical coherence tomography (OCT), full-field stimulus threshold (FST) test and clinical chemistry parameters were collected.[14]

VP-001 has PK and safety profiles favorable to an ocular therapeutic

The VP-001 half-life profile in NHPs supports an extended dosing interval in RP11 patients

PK

~20 days
Half-life in target cell layers that are affected in RP11

~100 days
Anticipated dosing interval*

Less frequent IVT dosing regimen
Preferred by patients

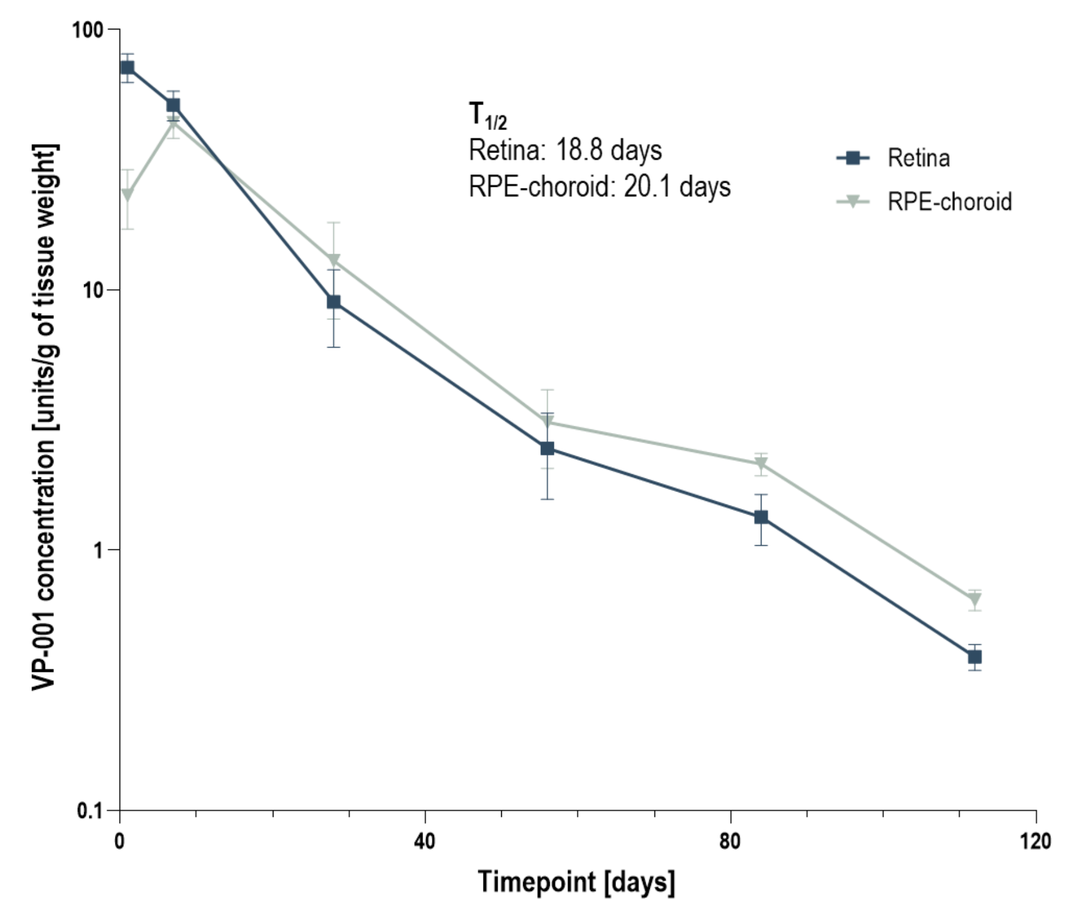

$T_{1/2}$
Retina: 18.8 days
RPE-choroid: 20.1 days
Retina
RPE-choroid
VP-001 concentration [units/g of tissue weight]
Timepoint [days]
100
10
1
0.1
0
40
80
120

VP-001 has a half-life of 19 and 20-days in the retina and RPE-choroid, respectively in non-human primates (NHP) after a single treatment, supporting a ~3-4 monthly dosing regimen in RP11 patients

GLP toxicology studies in NHPs demonstrate the safety and tolerability of VP-001 at clinically translatable doses

Dose of VP-001	µg/eye	# of eyes dosed	# of eyes with no adverse findings at 12-weeks
Control	0	12	12
Low	5	12	12
Medium	15	12	12
High	50	12	12

All doses tested in the GLP toxicology study in NHP were well-tolerated and safe throughout the 12-week study period

Platypus, a Phase 1 SAD FIH study shows encouraging safety and efficacy data in RP11

Safety/tolerability profile of VP-001 in the Phase 1 Single Ascending Dose (SAD) Study

- No Treatment Emergent-Serious Adverse Events (TE-SAEs) observed in any patient dosed with VP-001 to date
 - Including patients who have now received 3 doses of the highest dose (75 µg) of VP-001 in the SAD extension
- Treatment-Emergent Adverse Events (limited to patients who received lower doses) were mostly mild and procedure related
- No Treatment Emergent-Adverse Events of any nature in the highest dose cohort of the SAD study (after a single dose)

Both functional vision and visual function improvements relative to both control eye and baseline have been observed in RP11 patients treated with ≥ 30 µg of VP-001 in the Single Ascending Dose Study

*Results at Month 3 timepoint are used. If patient does not have data available at Month 3, data from Month 4 timepoint (if available) is used.

Figure 1. Improvements on mean retinal sensitivity have been observed in SAD cohort 3 and 4 patients treated with VP-001 at Month 3* following the first dose of VP-001 (n=4-6)

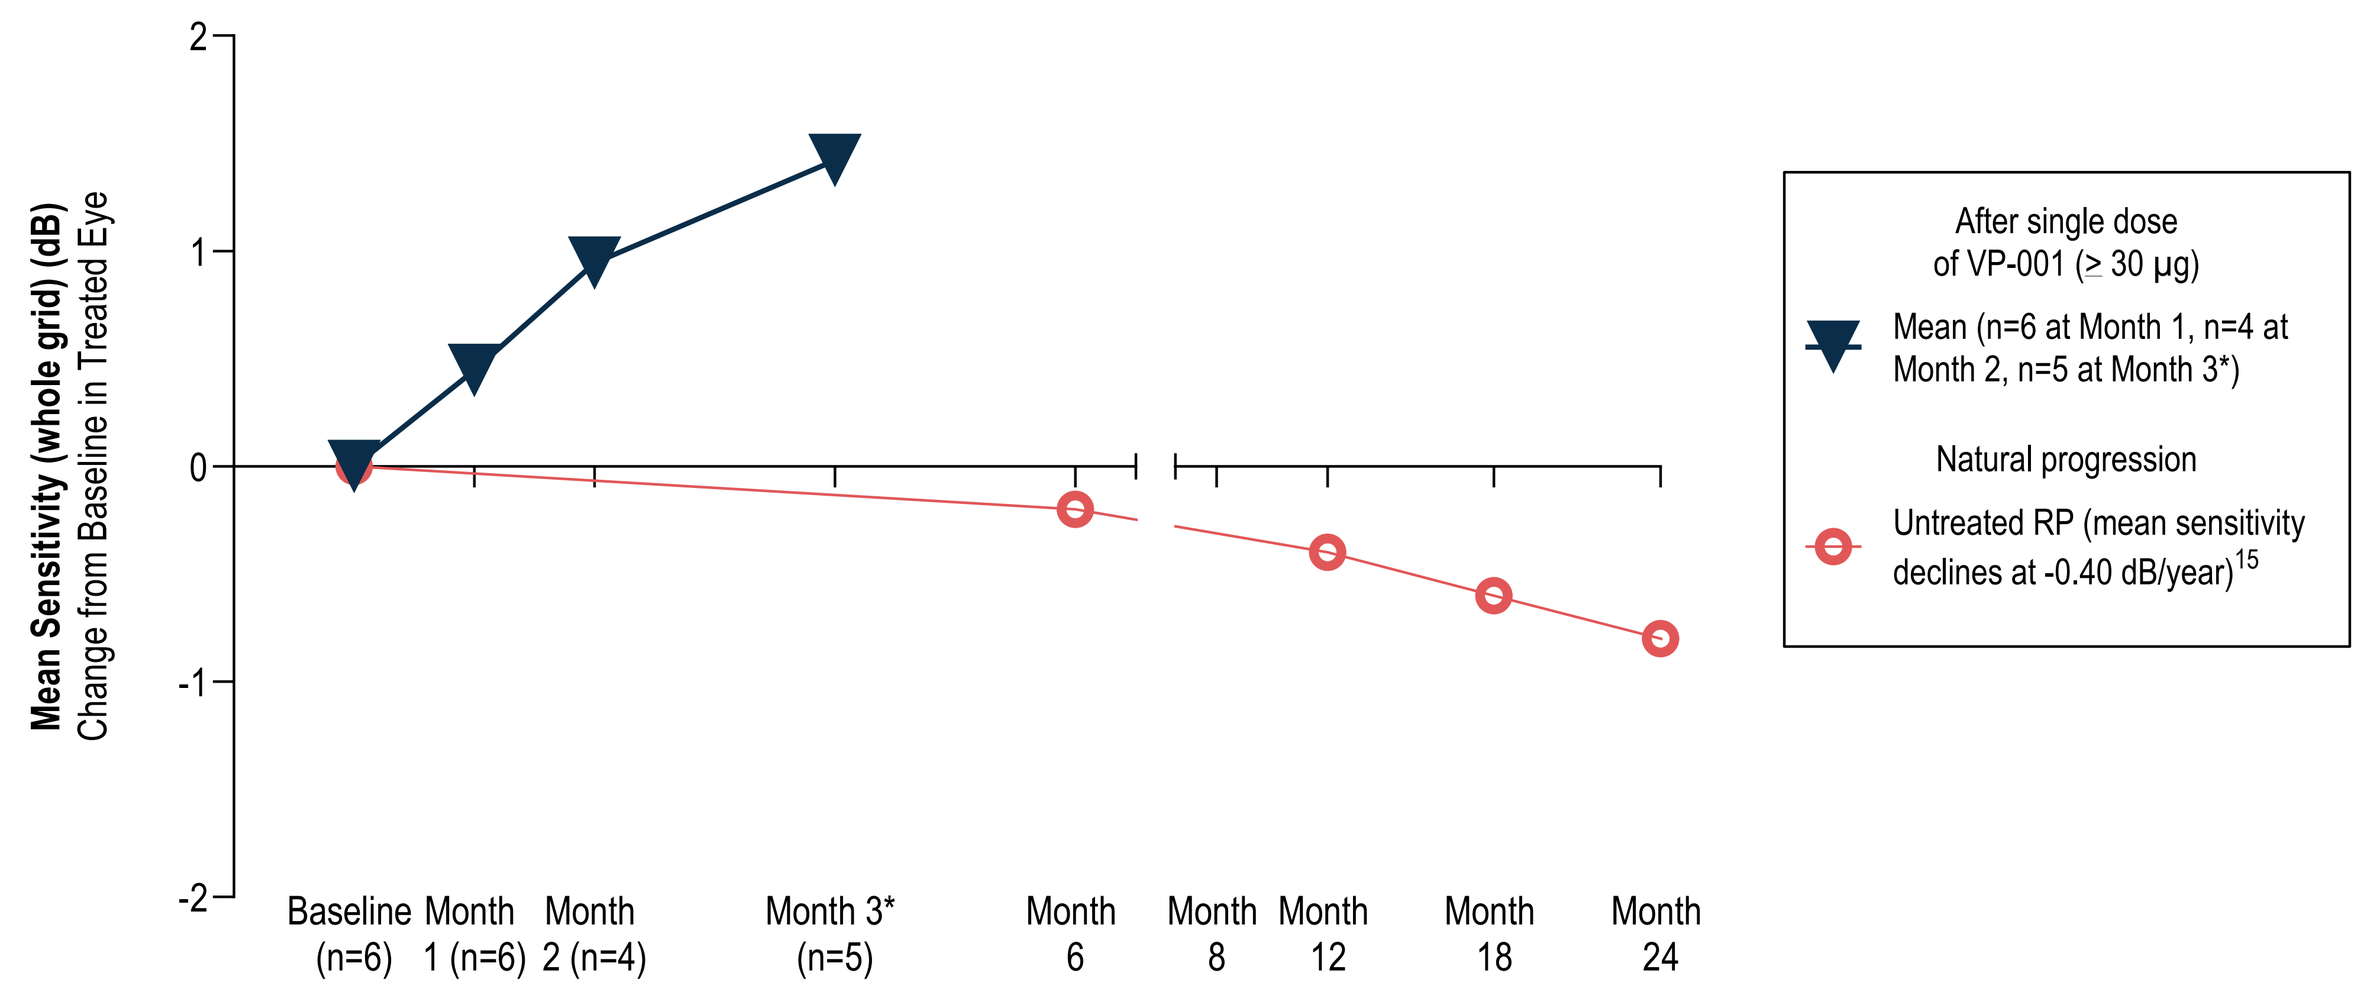

Mean Sensitivity (whole grid) (dB)
Change from Baseline in Treated Eye
2
1
0
-1
-2
Baseline (n=6)
Month 1 (n=6)
Month 2 (n=4)
Month 3* (n=5)
Month 6
Month 8
Month 12
Month 18
Month 24
After single dose of VP-001 (> 30 µg)
Mean (n=6 at Month 1, n=4 at Month 2, n=5 at Month 3*)
Natural progression
Untreated RP (mean sensitivity declines at -0.40 dB/year)15

Figure 2. Improvements (reductions) in the number of scotomas has been observed in SAD cohort 3 and 4 patients treated with VP-001 at Month 3* following the first dose of VP-001 (n=4-6)

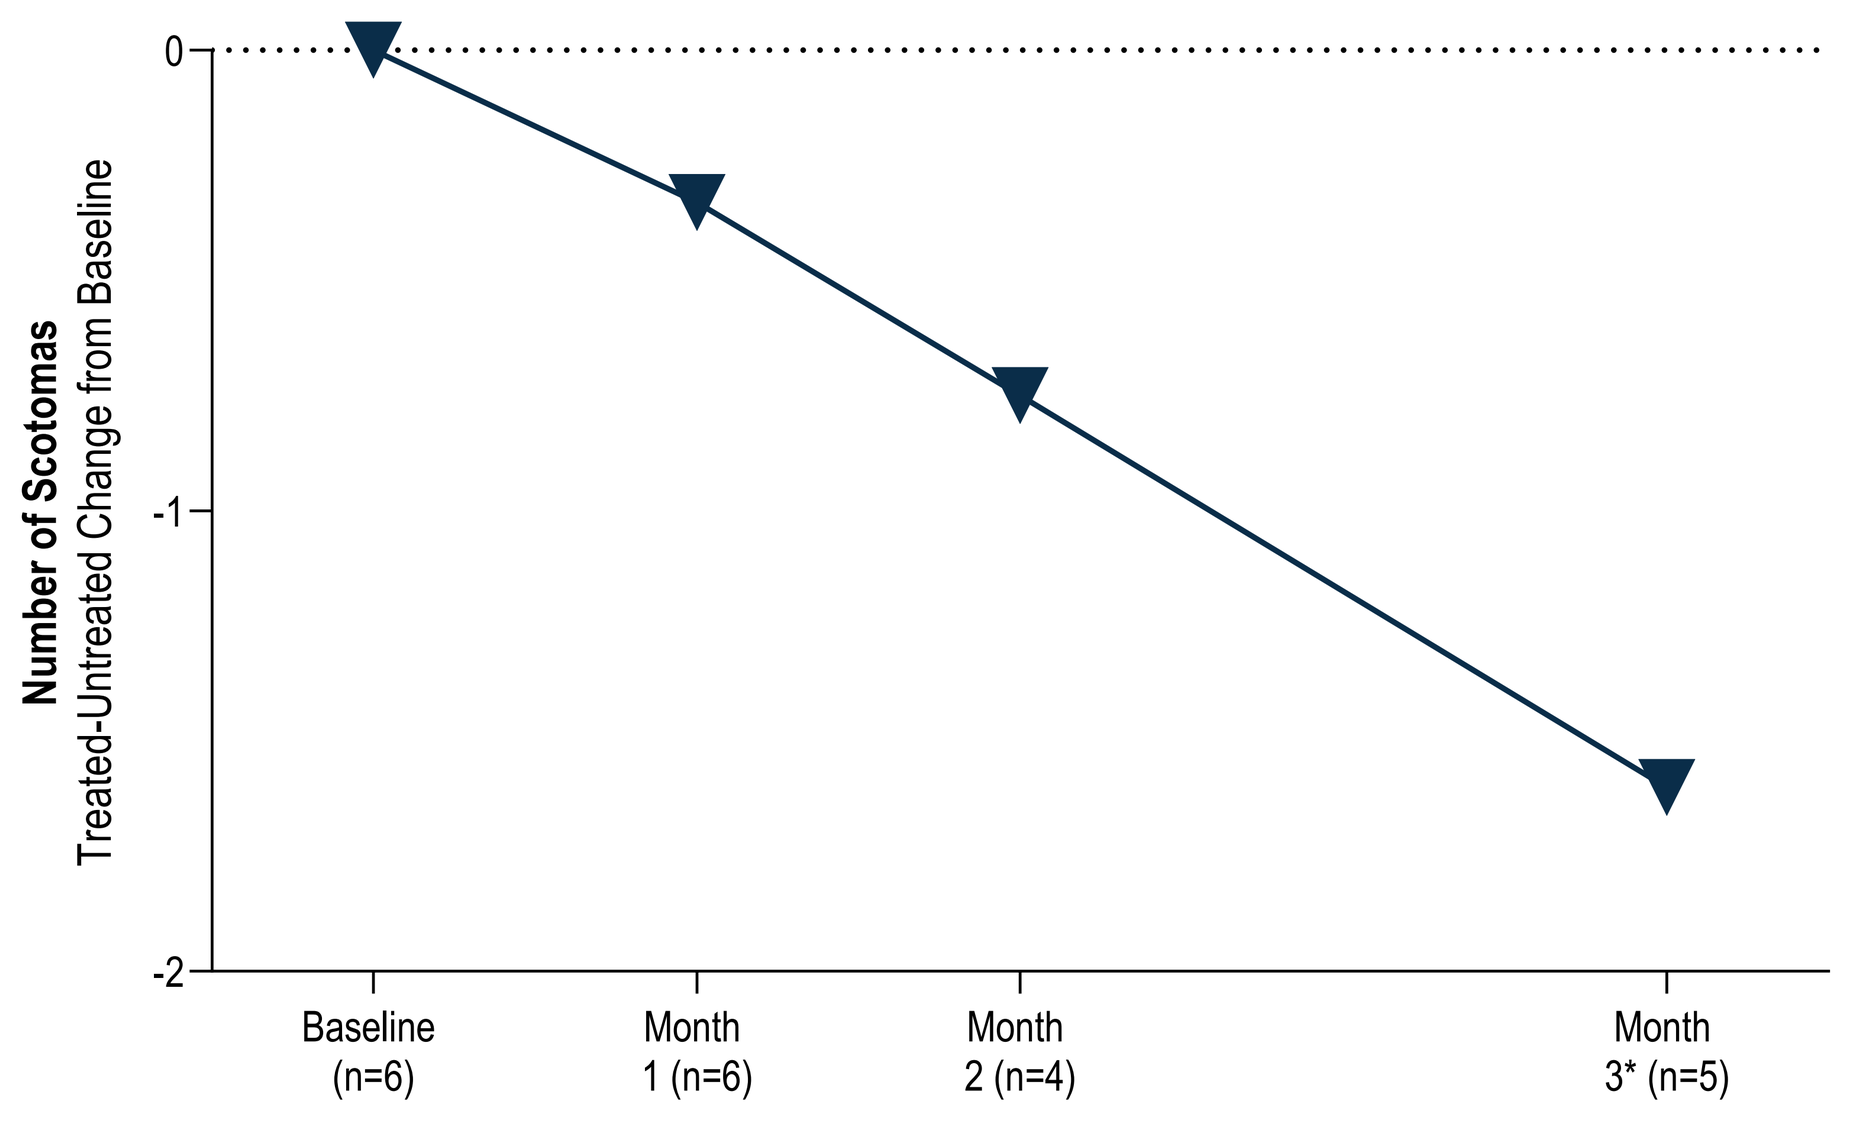

Number of Scotomas
Treated-Untreated Change from Baseline
0
-1
-2
Baseline (n=6)
Month 1 (n=6)
Month 2 (n=4)
Month 3* (n=5)

Figure 3. Improvements in Low-Luminance Visual Acuity has been observed in SAD cohort 3 and 4 patients treated with VP-001 at Month 3* following the first dose of VP-001 (n=4-6)

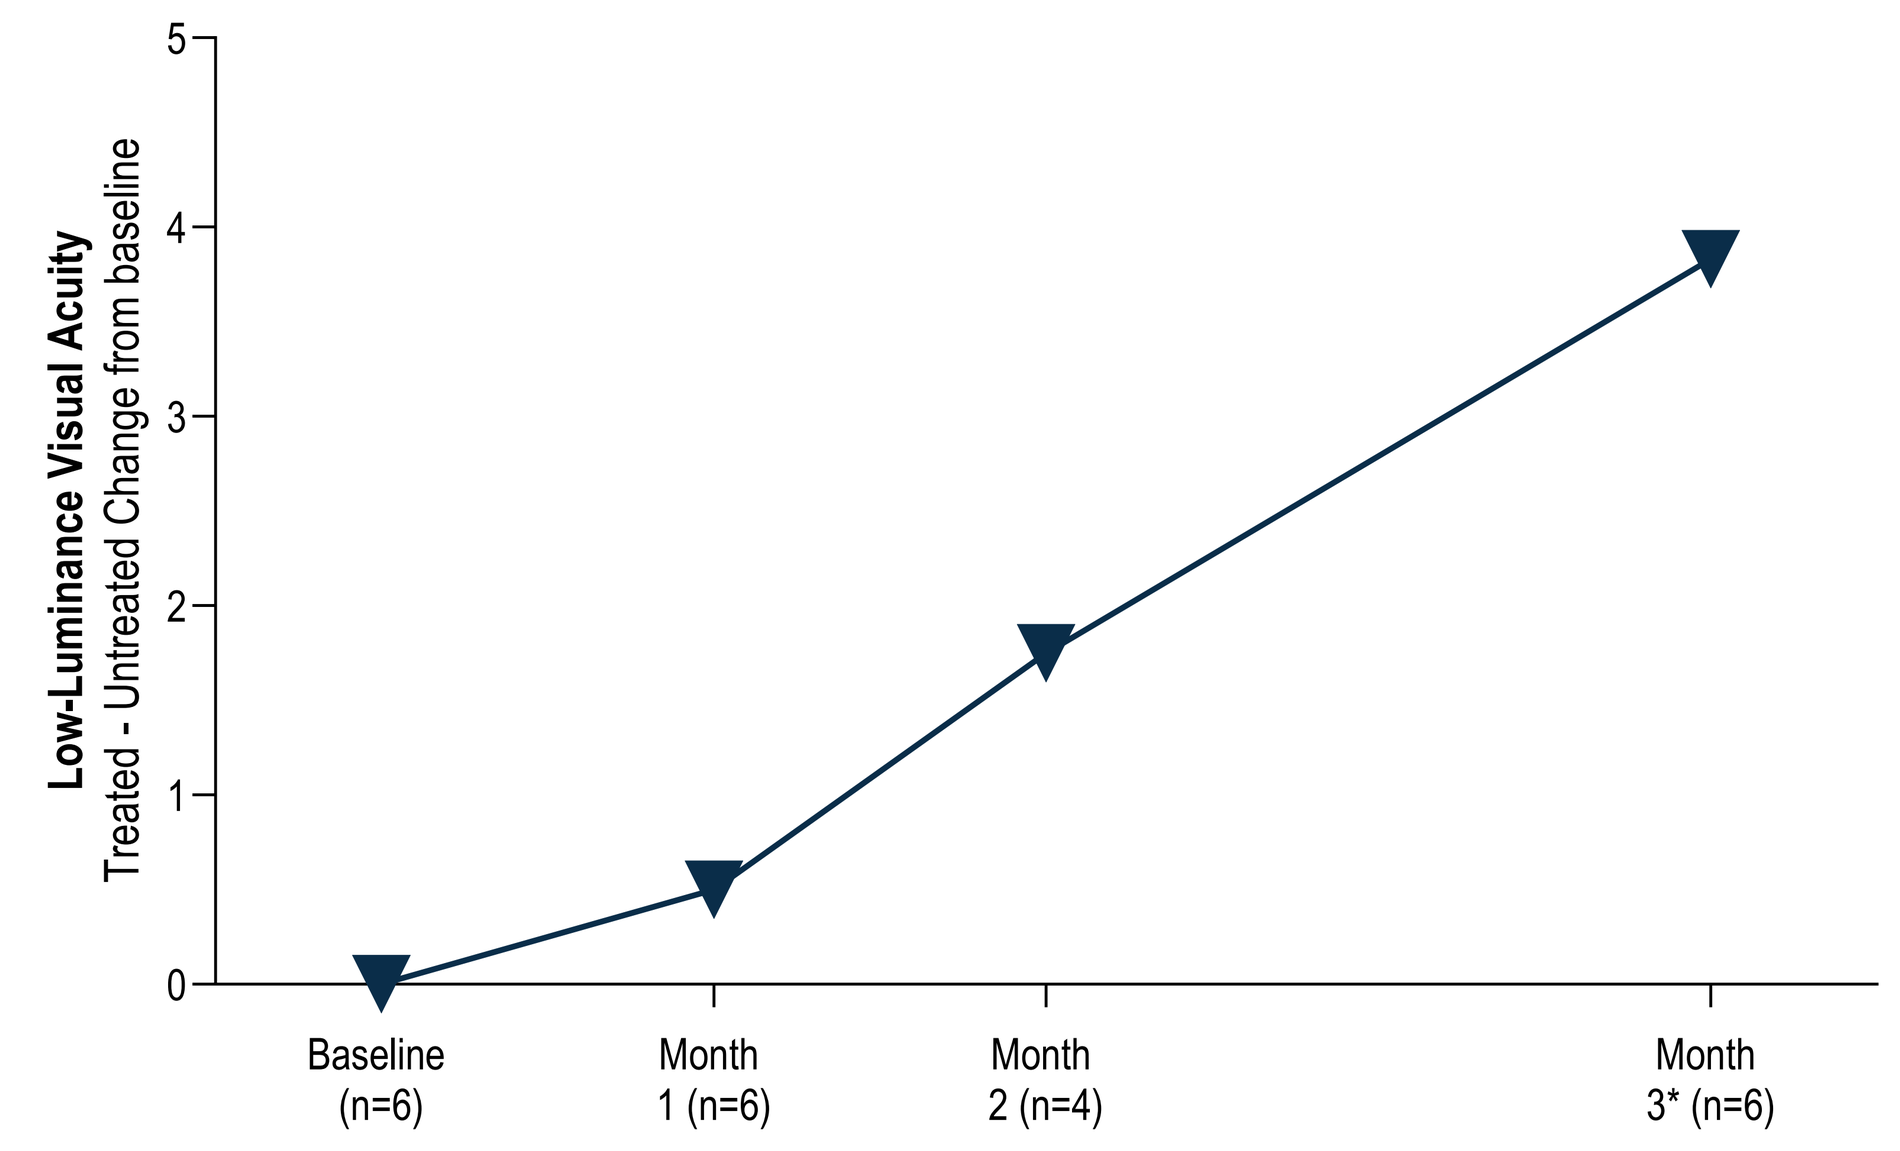

Low-Luminance Visual Acuity
Treated - Untreated Change from baseline
5
4
3
2
1
0
Baseline (n=6)
Month 1 (n=6)
Month 2 (n=4)
Month 3* (n=6)

Figure 4. Microperimetry heatmaps of an RP11 patient eye treated with a single 30 µg dose of VP-001

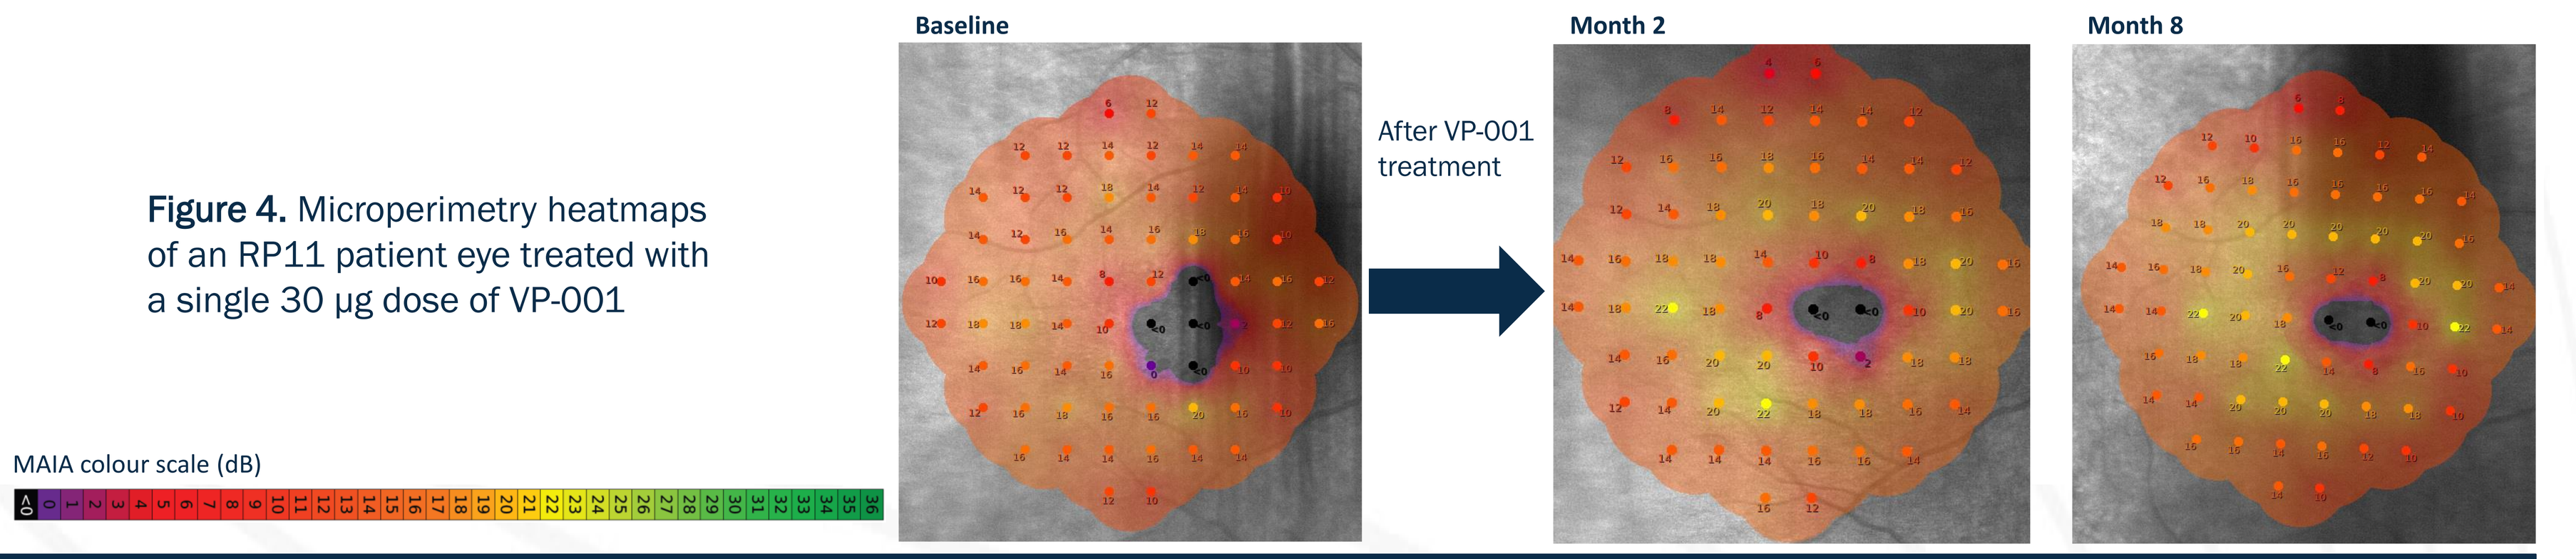

Baseline
After VP-001 treatment
Month 2
Month 8
MAIA colour scale (dB)

Clinical Path – VP-001 is the first drug candidate to have entered clinical trials in RP11

VP-001 is expected to progress into a registrational study in 2025 following data readouts from an ongoing Multiple Ascending Dose (MAD) study. The registrational study will be designed to support a potential New Drug Application in 2027*

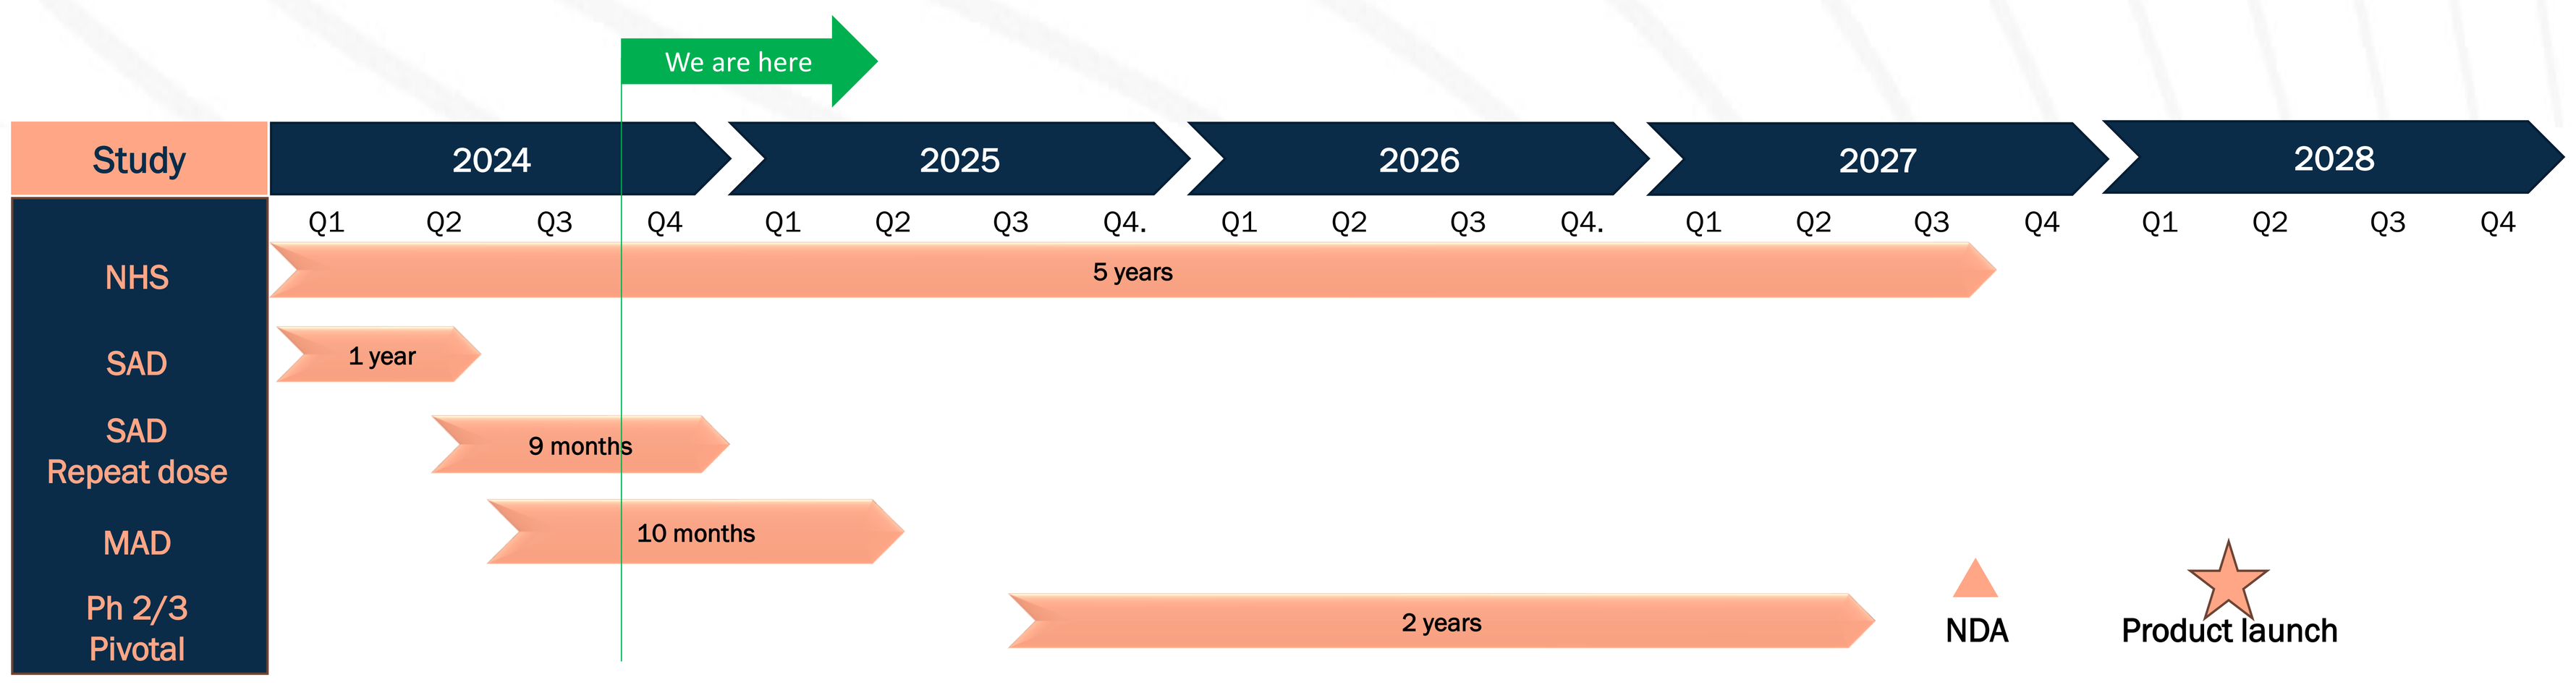

We are here
Study
2024
2025
2026
2027
2028
Q1
Q2
Q3
Q4
NHS
5 years
SAD
1 year
SAD Repeat dose
9 months
MAD
10 months
Ph 2/3 Pivotal
2 years
NDA
Product launch

*Based on management forecasts as at 28 October 2024 and subject to the risks set out in the company's ASX disclosures of 14 March 2024

Conclusions

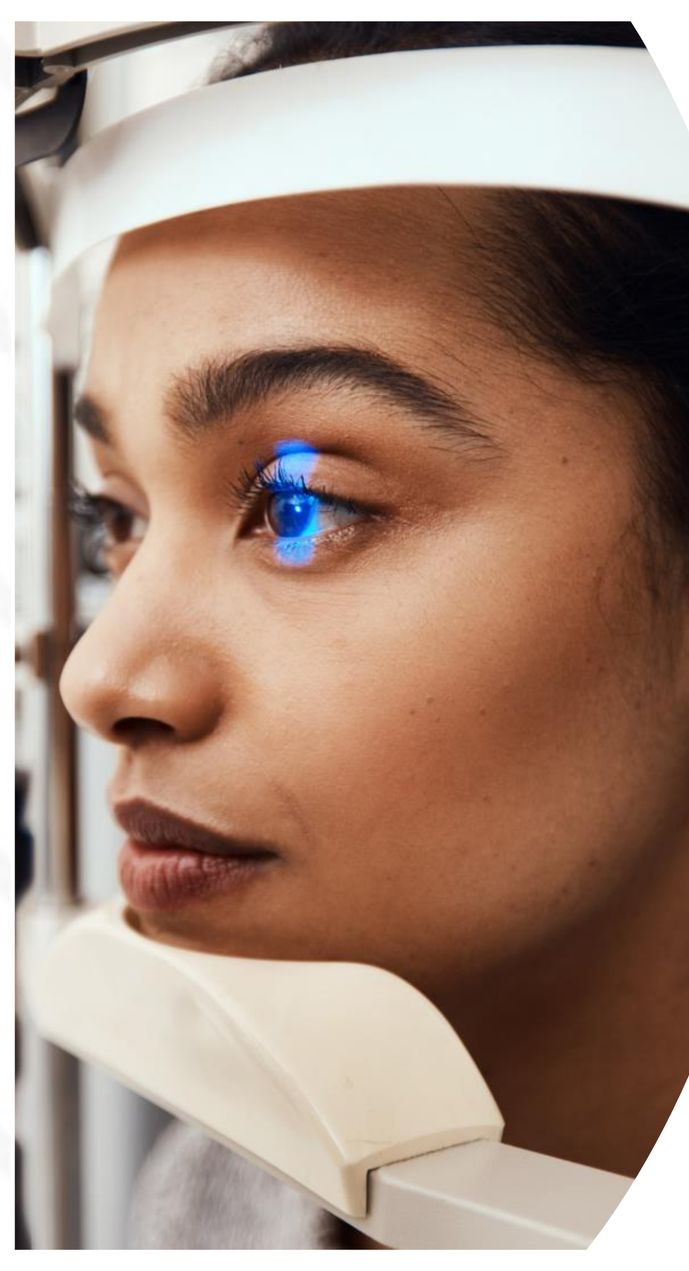

Preclinical and clinical findings support continued development of VP-001

- ✓ VP-001 is the first drug candidate to have entered clinical trials for Retinitis Pigmentosa type 11
- ✓ The results from Platypus SAD study demonstrate that single intravitreal injection of VP-001 is safe and well tolerated at 3, 10 and 30 and 75 µg doses
- ✓ No Treatment-Emergent Serious Adverse Events (TE-SAEs) were observed in any patient, including patients who have now received 3 doses of the highest dose (75 µg) of VP-001 in the SAD extension study
- ✓ Improvements in retinal sensitivity in VP-001 treated eyes were observed via microperimetry
- ✓ Improvements in Low-Luminance Visual Acuity in VP-001 treated eyes were observed
- ✓ VP-001 is expected to progress into a registrational study in 2025 following data readouts from an ongoing Multiple Ascending Dose (MAD) study

DisclosureBlock: Fred Chen, Code P (Patent) PYC Therapeutics, Code C (Consultant/Contractor) Novartis, Code C (Consultant/Contractor) Janssen, Code C (Consultant/Contractor) Bayer, Code C (Consultant/Contractor) Apellis, David G. Birch, Code C (Consultant/Contractor) PYC Therapeutics, Carla Jackson, Code E (Employment) PYC Therapeutics, Anna Mills, Code E (Employment) PYC Therapeutics, Laura Florez, Code E (Employment) PYC Therapeutics, Jessica Nichols, Code E (Employment) PYC Therapeutics, Paula Cunningham, Code E (Employment) PYC Therapeutics, Janya Grainok, Code E (Employment) PYC Therapeutics, Adam Martin, Code E (Employment) PYC Therapeutics, Subrata Das, Code E (Employment) PYC Therapeutics, Sue Fletcher, Code E (Employment) PYC Therapeutics, George Mitchell, Code E (Employment) PYC Therapeutics, Clare Guerrero, Code E (Employment) PYC Therapeutics, Sri Mudumba, Code E (Employment) PYC Therapeutics

References: **1.** Daiger, Bowne & Sullivan, *Cold Spring Harb Perspect Med* (2014). **2.** Daiger et al., *Adv Exp Med Biol* (2014). **3.** Martínez-Gimeno et al., *Invest Ophthalmol Vis Sci* (2003). **4.** Audo et al., *BMC Med Genet* (2010). **5.** Coussa et al., *Invest Ophthalmol Vis Sci* (2015). **6.** Lim et al., *Arch Ophthalmol* (2009). **7.** Martin-Merida et al., *Invest Ophthalmol Vis Sci* (2018). **8.** Sullivan et al., *Invest Ophthalmol Vis Sci* (2006). **9.** Sullivan et al., *Invest Ophthalmol Vis Sci* (2013). **10.** Van Cauwenbergh et al., *PLoS One* (2017). **11.** Xu et al., *Mol Vis* (2012). **12.** Szustakowski et al., *Medrxiv* (2020). **13.** Venturini et al. *PLOS Genetics* (2012). **14.** Refer ASX announcements of 7 October 2020, 16 December 2020, 10 May 2022, 7 November 2022, 6 May 2024 and 30 August 2024 **15.** Iftikhar M, et al. Progression of Retinitis Pigmentosa as Measured on Microperimetry: The PREP-1 Study. Ophthalmol Retina. 2018;2(5):502-7

Illustrations created with Biorender.com